COPENHAGEN
21ST CONGRESS
JUNE 9 – 12 | 2016
European Hematology Association

Development of a novel first-in-class oral inhibitor of the NOTCH pathway

R.Lehal[1] , V.Frismantas[2], V. Reinmuller[3] , G.Turcatti[4] , J-P. Bourquin[2] , F.Radtke[3]

1. Cellestia Biotech AG, Basel, Switzerland
2. University Children's Hospital, Zurich, Switzerland
3. Swiss Institute for Experimental Cancer Research, EPFL, Switzerland
4. Biomolecular Screening Facility, EPFL, Lausanne, Switzerland

CELLESTIA BIOTECH
INNOVATIVE THERAPEUTICS THAT MATTER

BACKGROUND

NOTCH signaling is a developmental pathway known to play critical roles during embryonic development as well as for the regulation of self-renewing tissues. Aberrant activation of NOTCH signaling leads to deregulation of the self-renewal process resulting in sustained proliferation, evasion of cell death, loss of differentiation capacity, invasion and metastasis, all of which are hallmarks of cancer. Over activation of NOTCH in human cancers can be a consequence of over expression of NOTCH ligands/receptors, GOF mutations in NOTCH receptors as well as chromosomal translocations leading to constitutive activation of the pathway.

OBJECTIVES

Two different therapeutic approaches have been utilized to block NOTCH; a) monoclonal blocking antibodies (Mabs) against NOTCH receptors/ligands and b) small molecule γ-secretase inhibitors (GSIs). However, these approaches can only be effective if cancer cells express full length NOTCH ligands/receptors. In human cancers harbouring NOTCH gene fusions due to chromosomal translocations, the use of Mabs and GSIs will have limited clinical benefit. A third, yet not fully explored approach could be the direct targeting of NOTCH transcriptional activation complex using small molecule inhibitors. The aim of this study was to discover and develop small molecule inhibitors of the NOTCH pathway able to block NOTCH signalling by targeting NOTCH transcription complex

RESULTS

Figure 1: CB-103 acts as a Pan-NOTCH inhibitor by targeting NOTCH transcriptional activation complex.

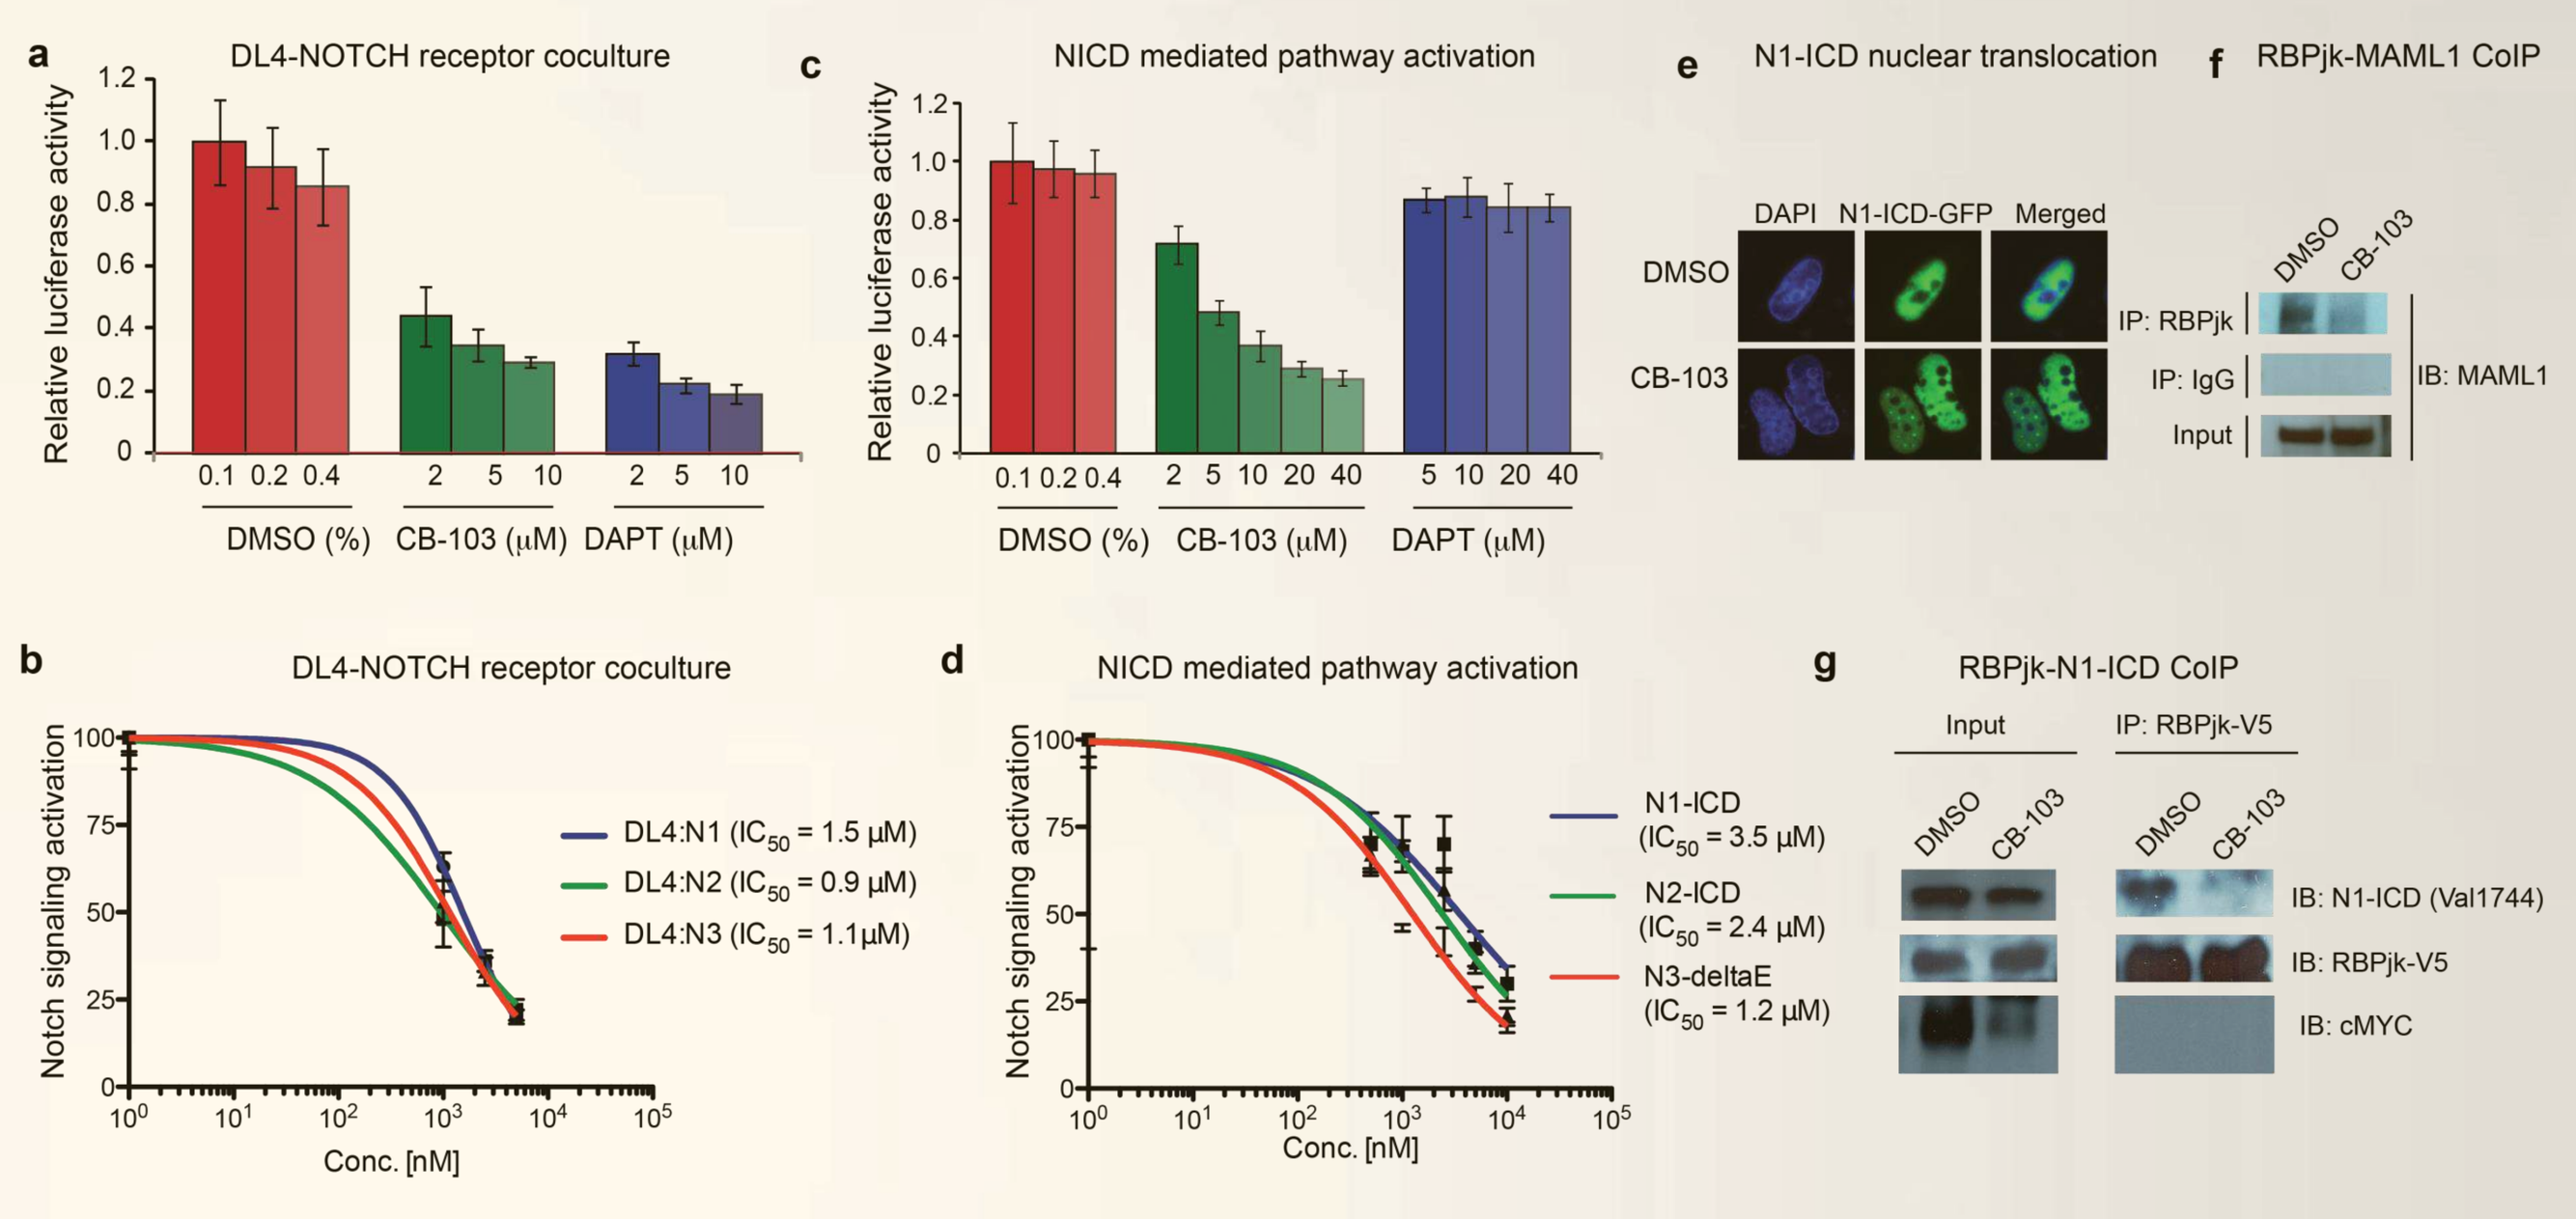

a DL4-NOTCH receptor coculture
Relative luciferase activity
DMSO (%) CB-103 (µM) DAPT (µM)
b DL4-NOTCH receptor coculture
Notch signaling activation
DL4:N1 (IC_{50} = 1.5 µM)
DL4:N2 (IC_{50} = 0.9 µM)
DL4:N3 (IC_{50} = 1.1µM)
Conc. [nM]
c NICD mediated pathway activation
d NICD mediated pathway activation
N1-ICD (IC_{50} = 3.5 µM)
N2-ICD (IC_{50} = 2.4 µM)
N3-deltaE (IC_{50} = 1.2 µM)
e N1-ICD nuclear translocation
DAPI N1-ICD-GFP Merged
DMSO
CB-103
f RBPjk-MAML1 CoIP
IP: RBPjk
IP: IgG
Input
IB: MAML1
g RBPjk-N1-ICD CoIP
Input IP: RBPjk-V5
IB: N1-ICD (Val1744)
IB: RBPjk-V5
IB: cMYC

Figure 2: CB-103 blocks NOTCH signaling in human T-ALL leukemic cell lines.

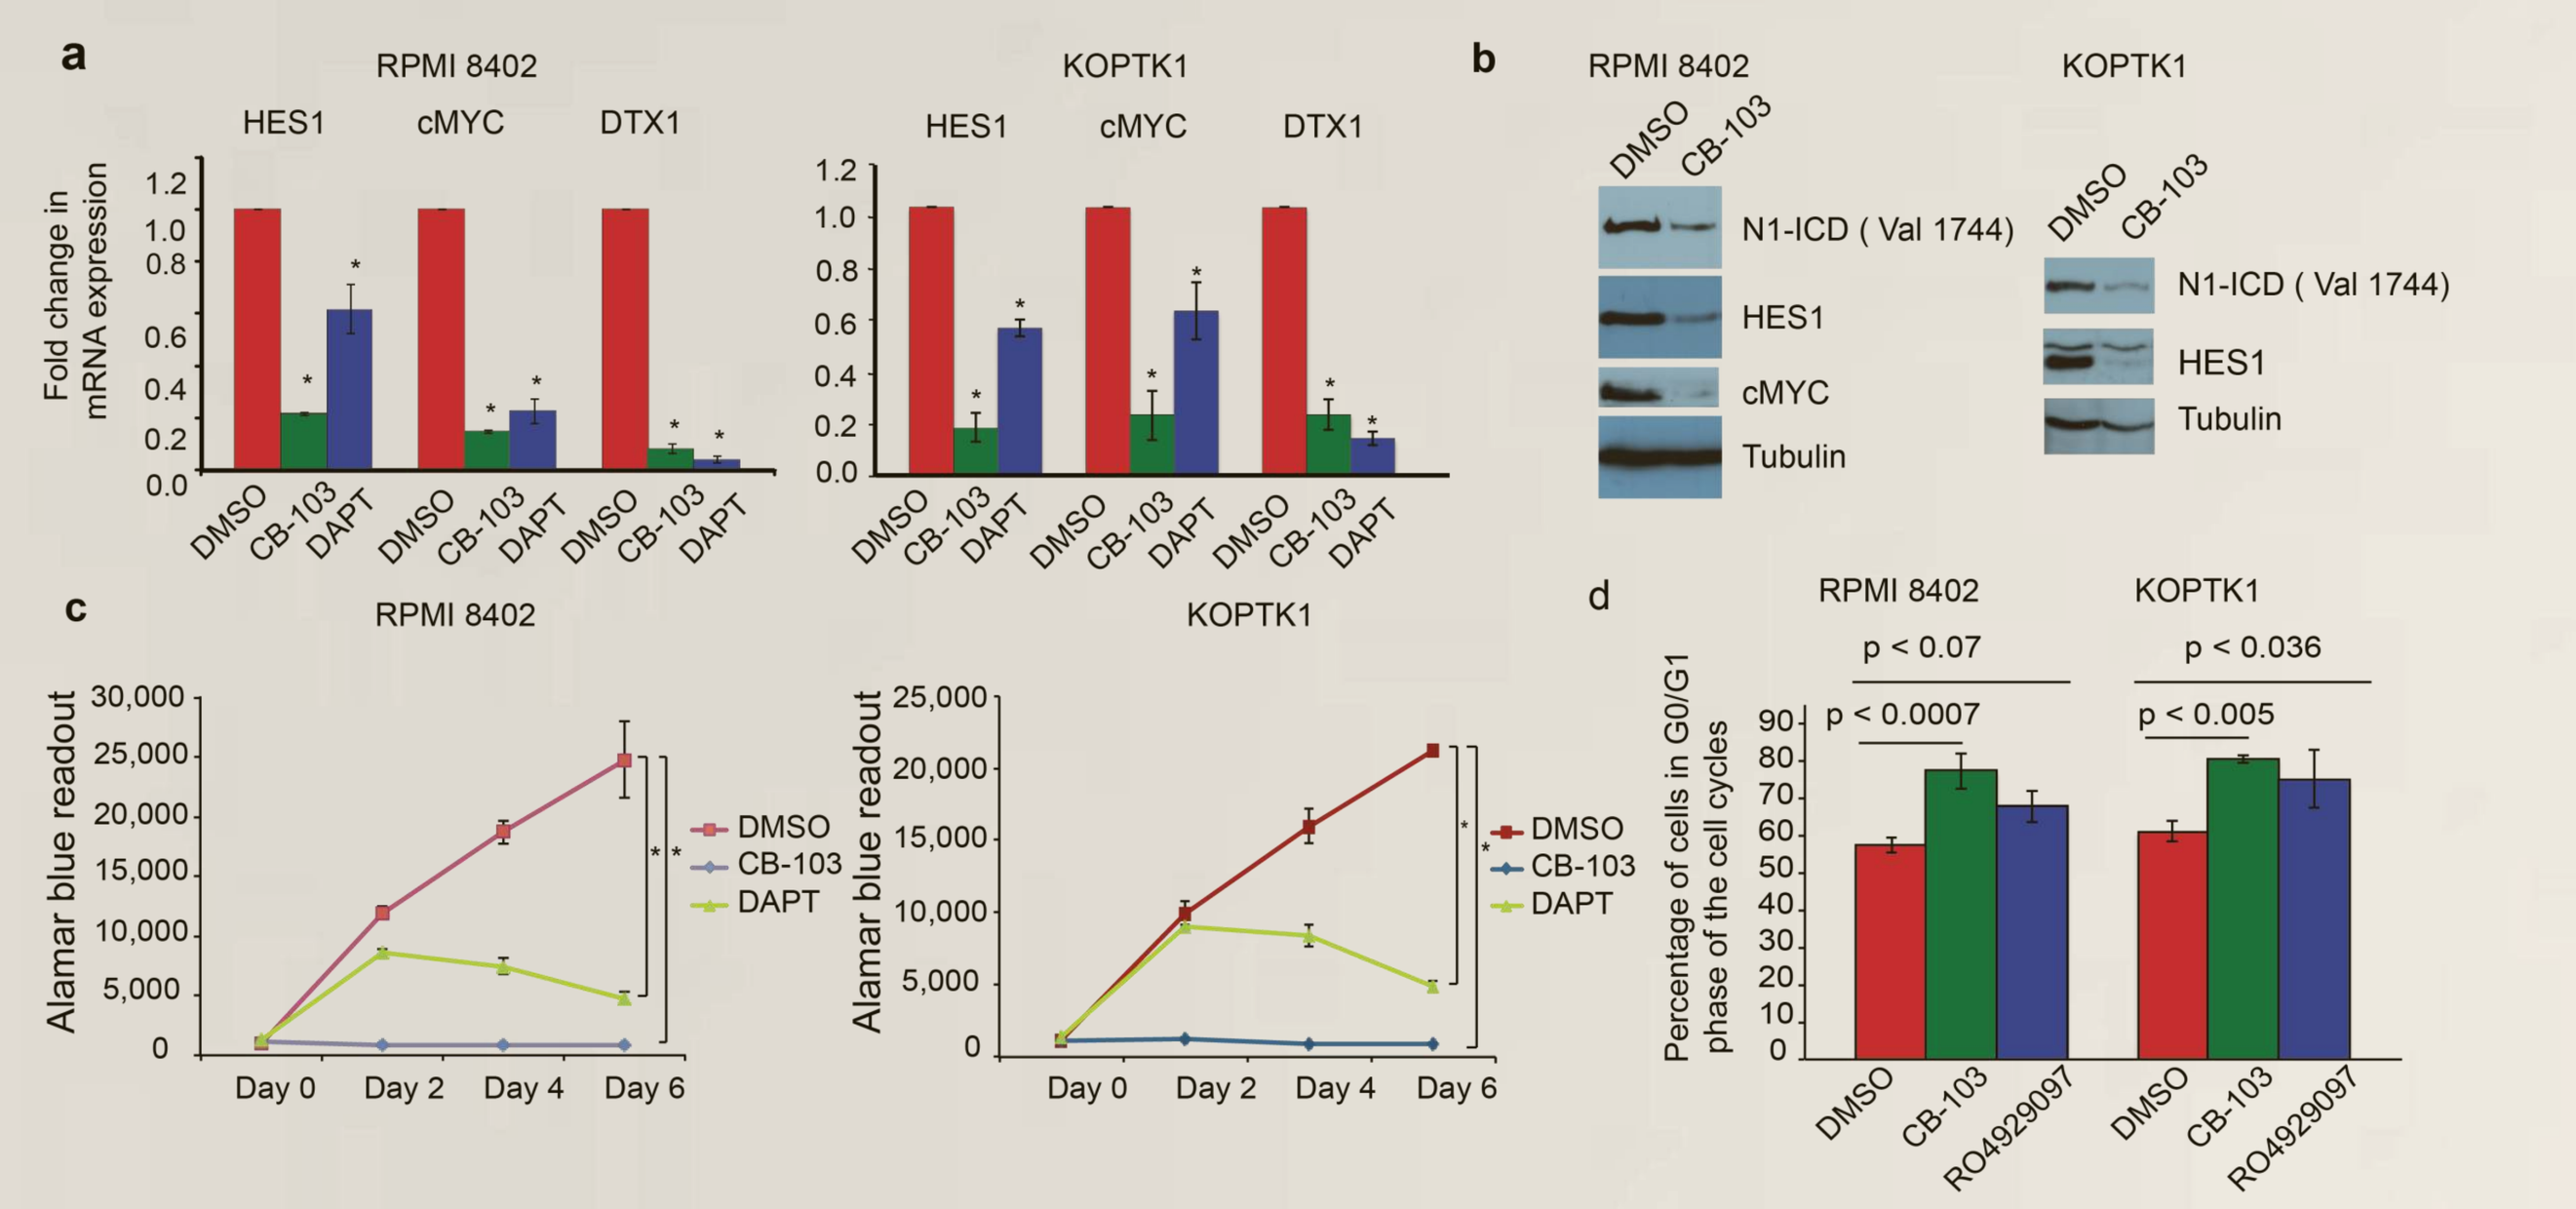

a RPMI 8402 KOPTK1
HES1 cMYC DTX1
Fold change in mRNA expression
DMSO CB-103 DAPT
b RPMI 8402 KOPTK1
N1-ICD (Val 1744)
HES1
cMYC
Tubulin
c RPMI 8402 KOPTK1
Alamar blue readout
Day 0 Day 2 Day 4 Day 6
DMSO
CB-103
DAPT
d RPMI 8402 KOPTK1
p < 0.07
p < 0.0007
p < 0.036
p < 0.005
Percentage of cells in G0/G1 phase of the cell cycles
DMSO CB-103 RO4929097

Figure 3: CB-103 inhibits NOTCH in GSI/Mabs resistant human cancer cell lines.

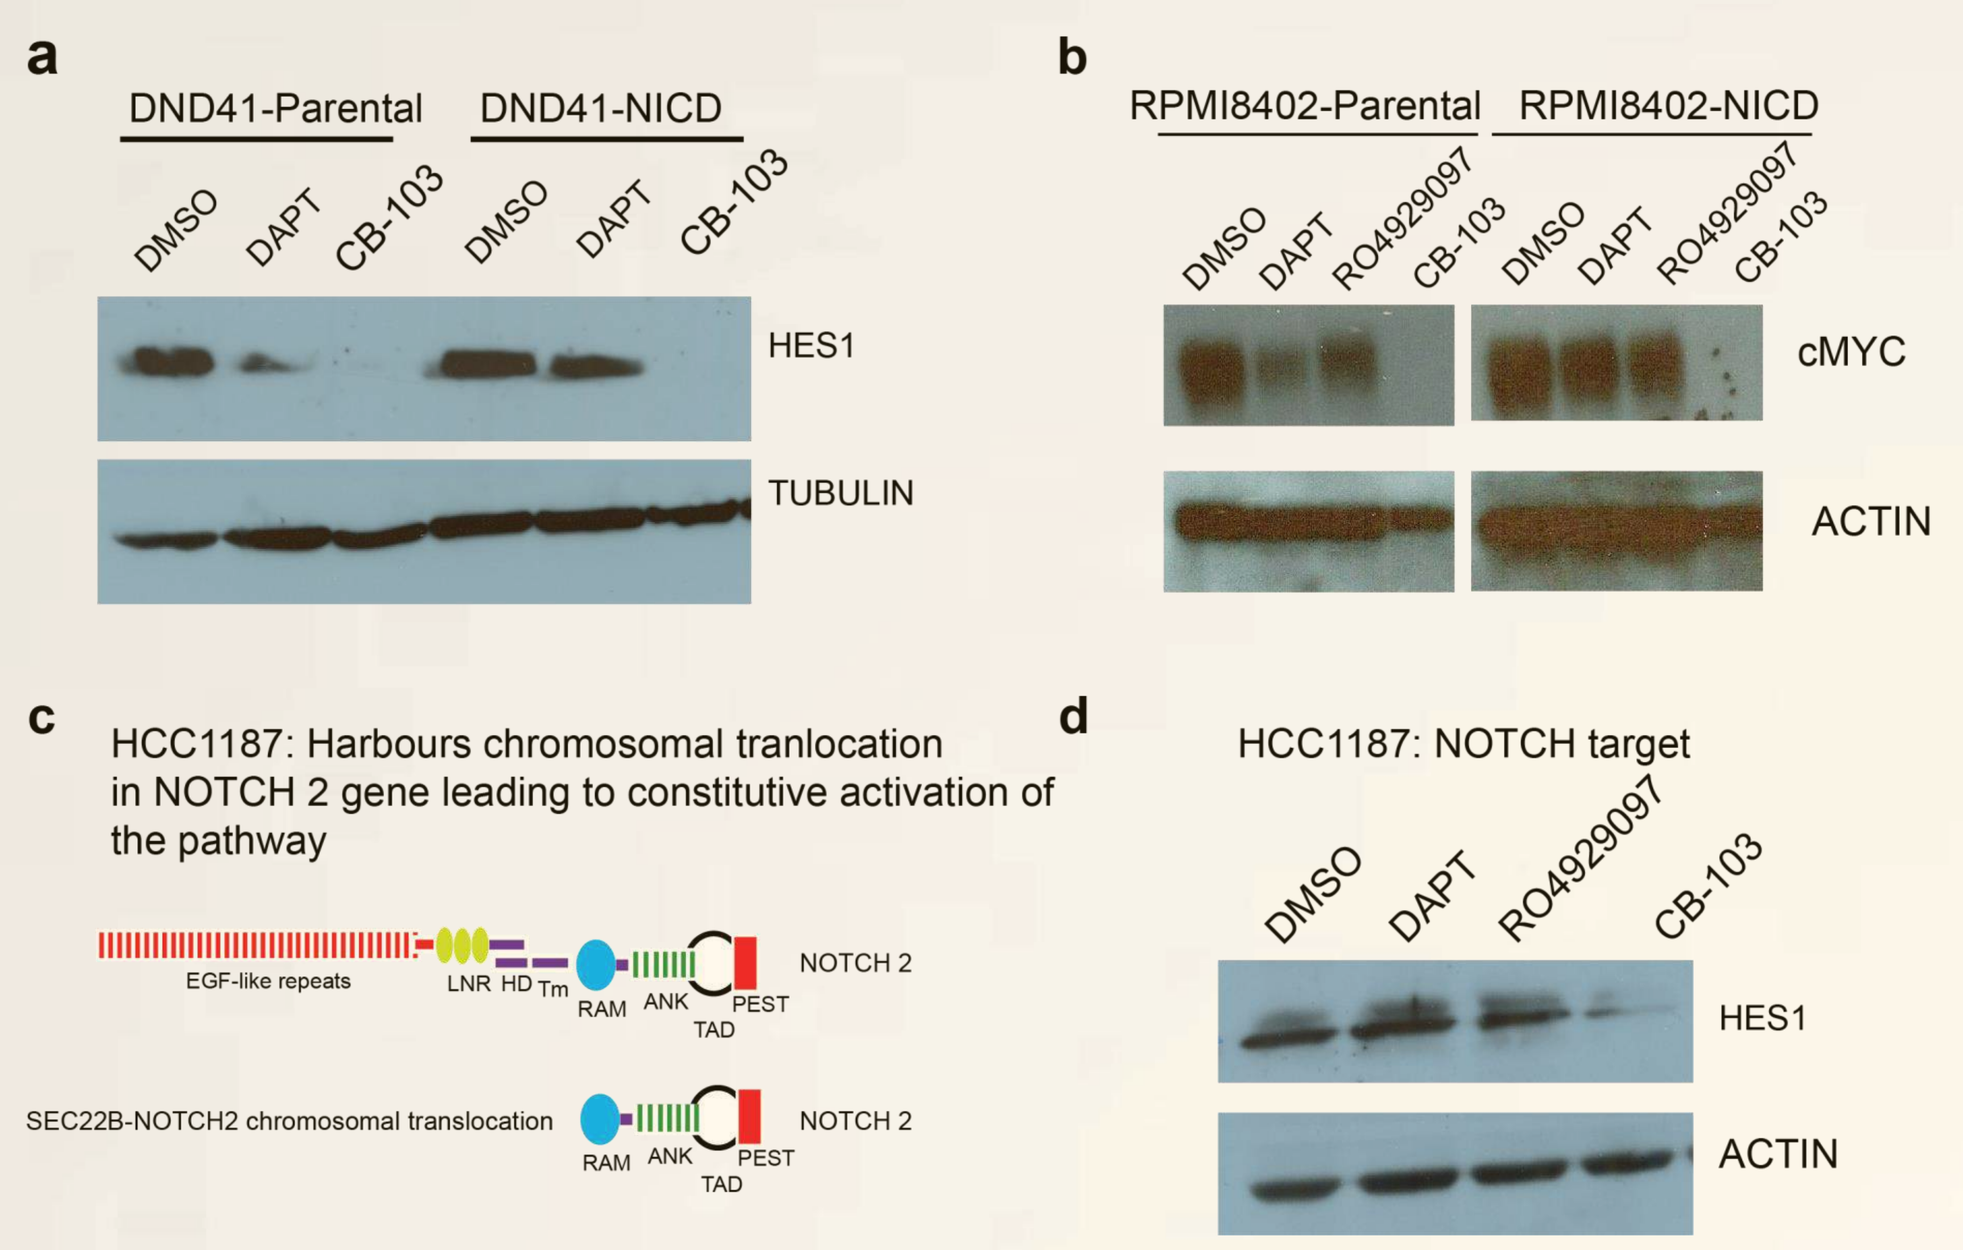

a DND41-Parental DND41-NICD
DMSO DAPT CB-103
HES1
TUBULIN
b RPMI8402-Parental RPMI8402-NICD
DMSO DAPT RO4929097 CB-103
cMYC
ACTIN
c HCC1187: Harbours chromosomal tranlocation in NOTCH 2 gene leading to constitutive activation of the pathway
EGF-like repeats LNR HD Tm RAM ANK TAD PEST NOTCH 2
SEC22B-NOTCH2 chromosomal translocation
d HCC1187: NOTCH target
HES1
ACTIN

Figure 4: CB-103 exhibits *in vivo* activity and blocks NOTCH dependent cellular processes.

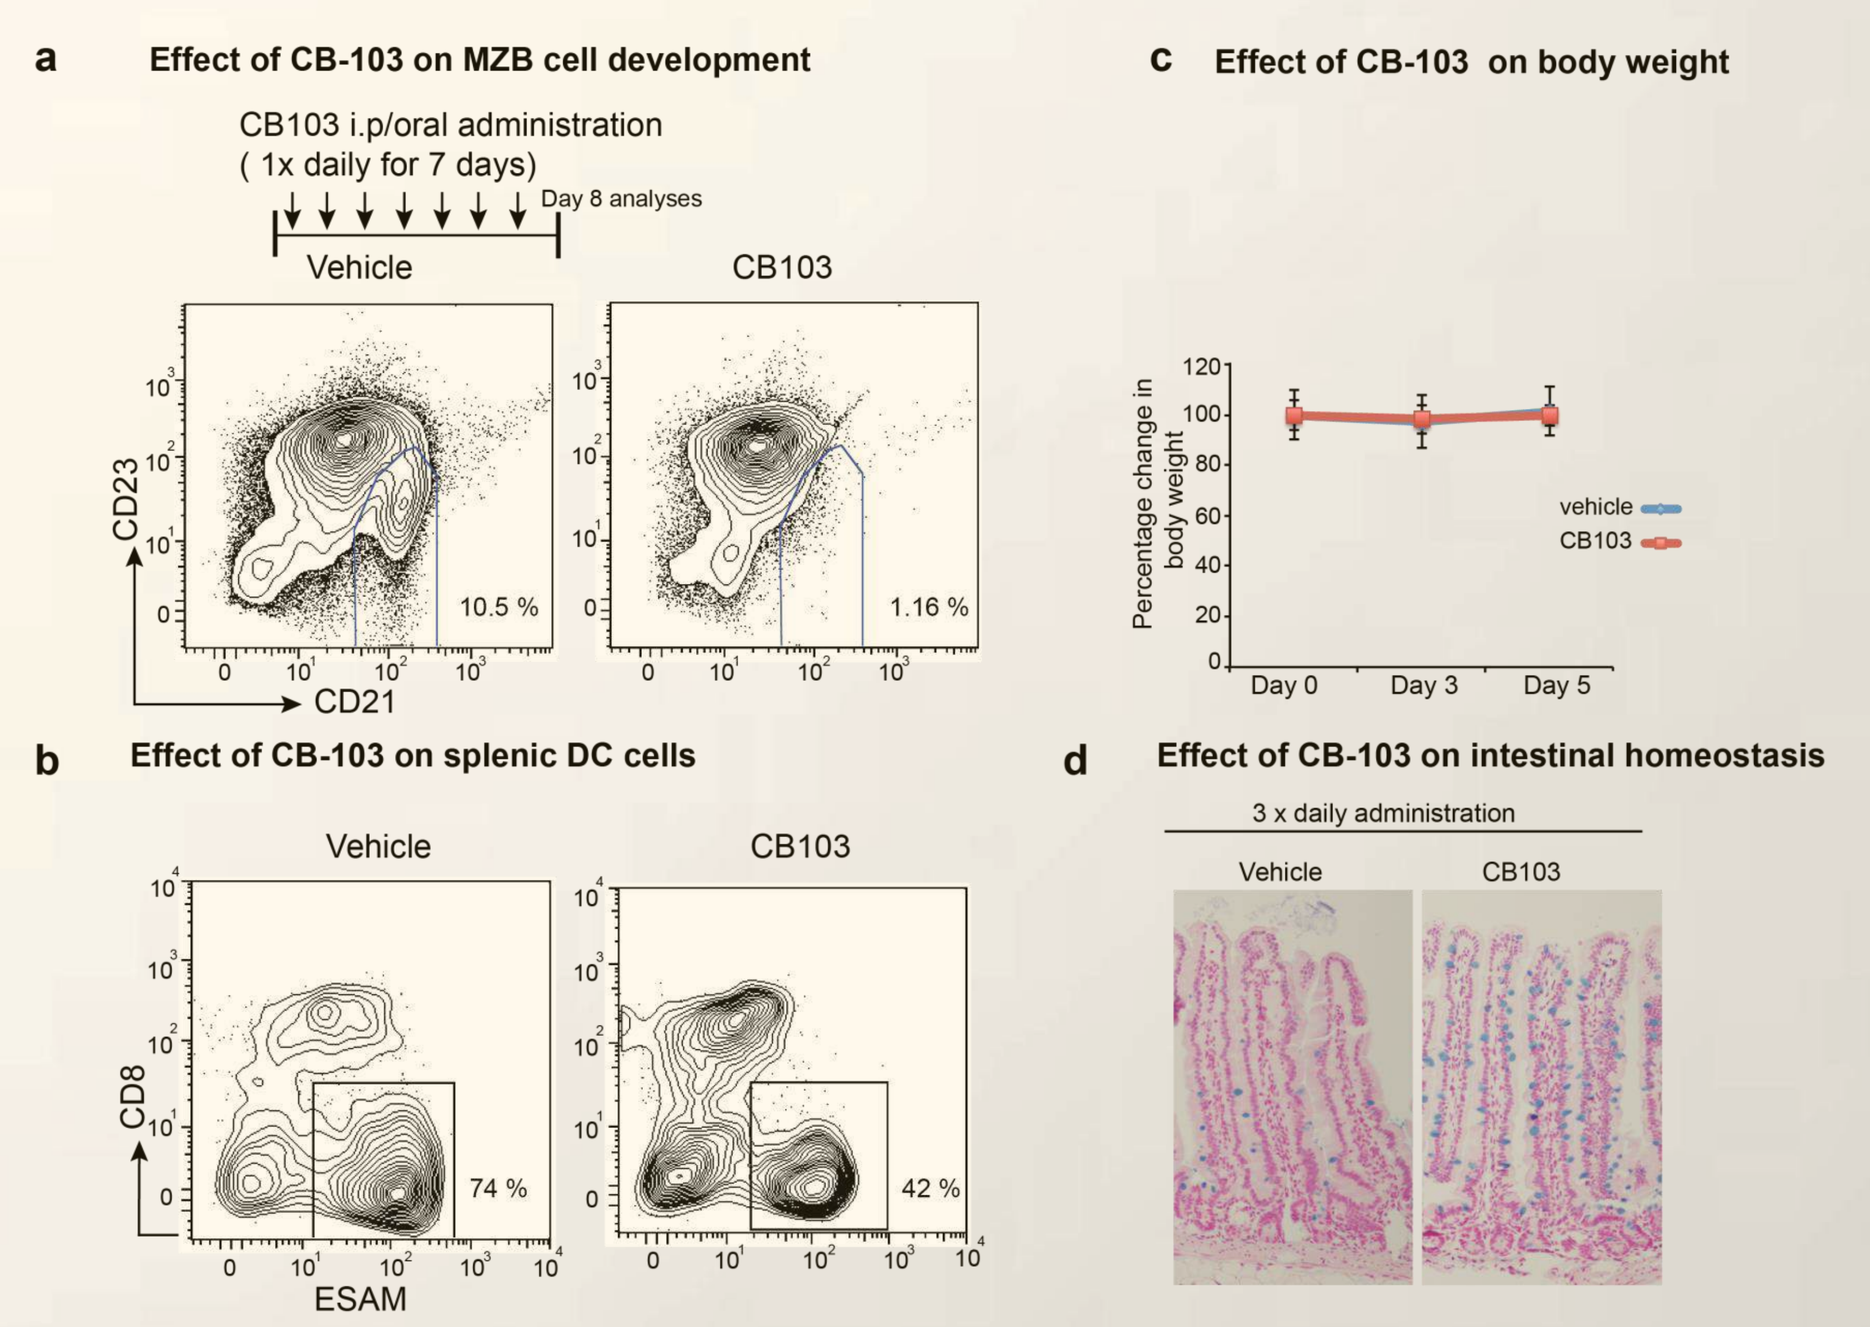

a Effect of CB-103 on MZB cell development
CB103 i.p/oral administration
(1x daily for 7 days)
Day 8 analyses
Vehicle CB103
CD23
CD21
10.5 %
1.16 %
b Effect of CB-103 on splenic DC cells
CD8
ESAM
74 %
42 %
c Effect of CB-103 on body weight
Percentage change in body weight
Day 0 Day 3 Day 5
vehicle
CB103
d Effect of CB-103 on intestinal homeostasis
3 x daily administration
Vehicle CB103

Figure 5: CB-103 impedes progression of NOTCH dependent xenograft model of human leukemia.

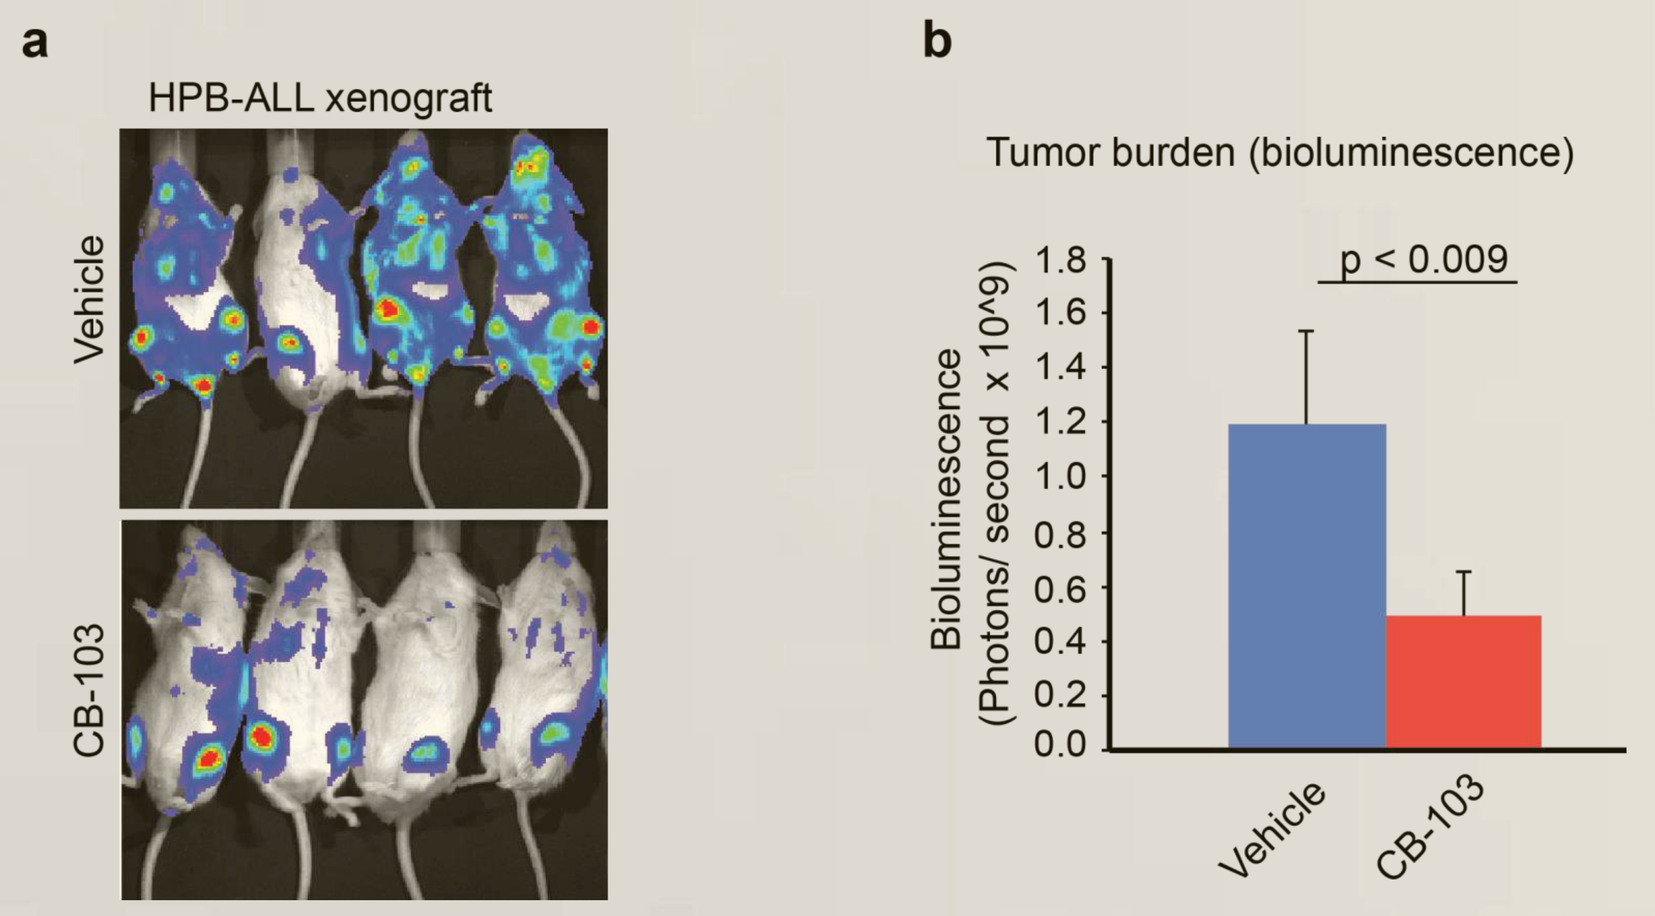

a HPB-ALL xenograft
Vehicle
CB-103
b Tumor burden (bioluminescence)
p < 0.009
Bioluminescence (Photons/ second x 10^9)
Vehicle CB-103

Figure 6: CB-103 blocks *in vivo* growth of PDX models of T-ALL

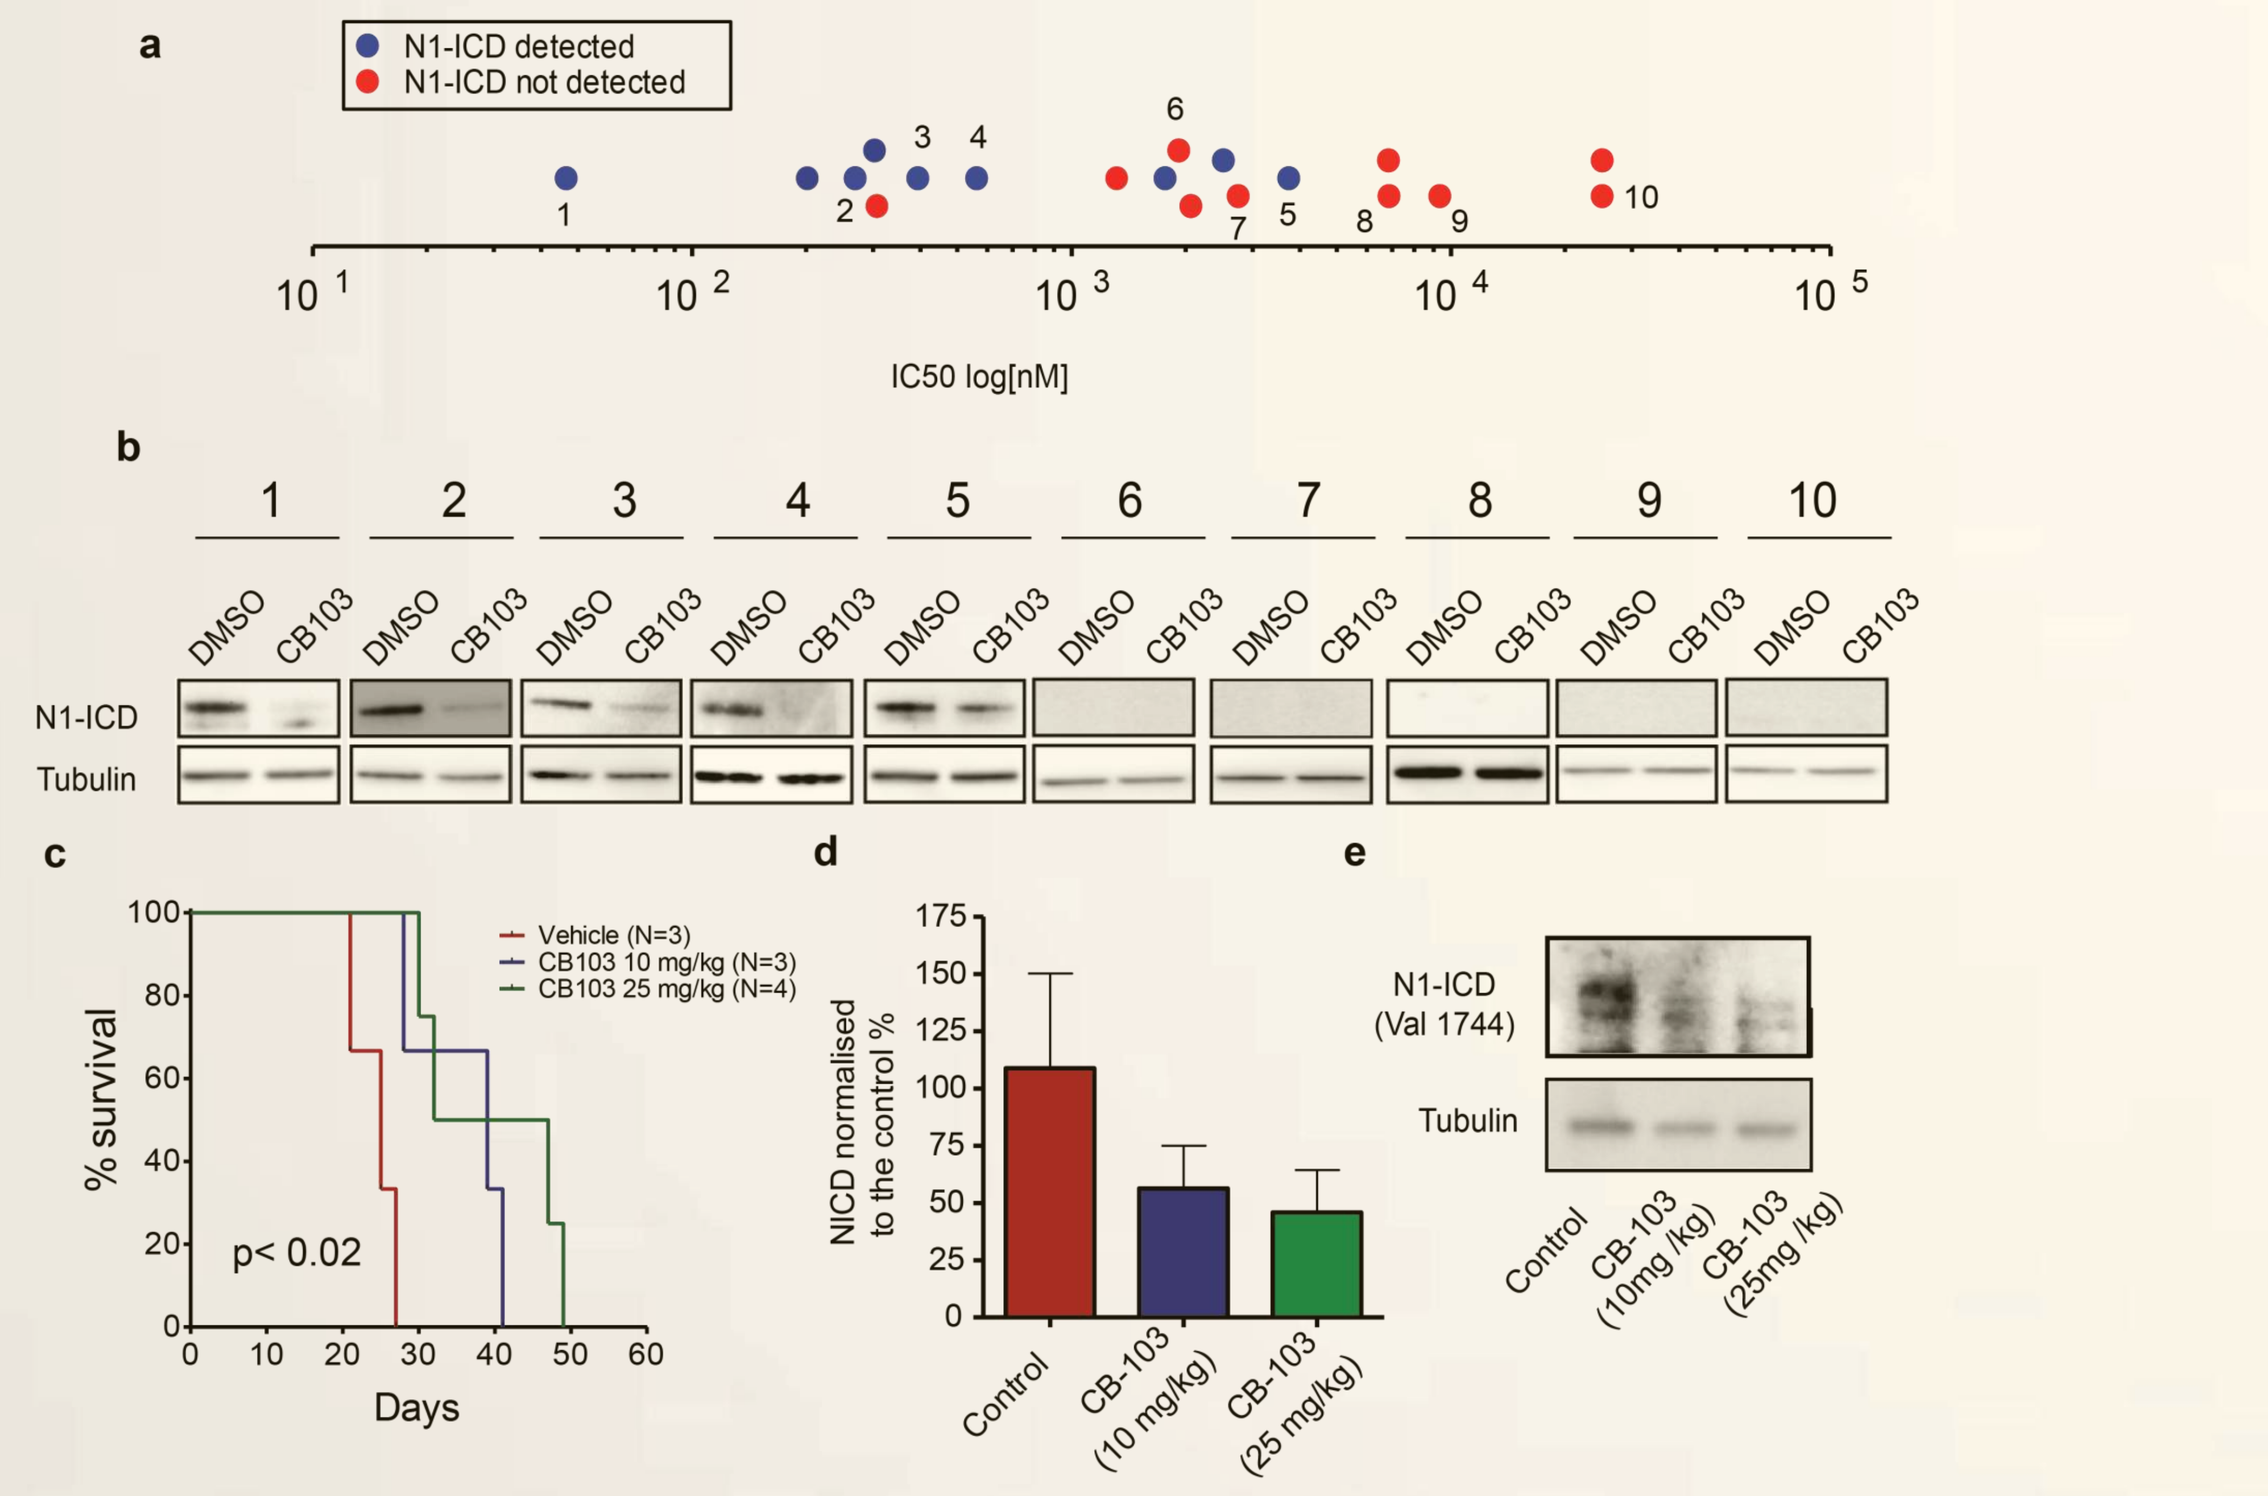

a N1-ICD detected
N1-ICD not detected
IC50 log[nM]
b N1-ICD
Tubulin
DMSO CB103
c % survival
Days
p< 0.02
Vehicle (N=3)
CB103 10 mg/kg (N=3)
CB103 25 mg/kg (N=4)
d NICD normalised to the control %
Control CB-103 (10 mg/kg) CB-103 (25 mg/kg)
e N1-ICD (Val 1744)
Tubulin
Control CB-103 (10mg /kg) CB-103 (25mg /kg)

MATERIALS & METHODS

We have developed a HTS compatible DL4-N1 coculture and NICD-driven NOTCH responsive reporter system that can be utilized to screen libraries of small molecules. Using this platform we have discovered and identified a novel, orally-active small molecule inhibitor, named CB-103, of the NOTCH pathway. CB-103 blocks NOTCH signaling by directly targeting the NOTCH transcriptional activation complex.

We have utilized several biochemical and cell-based assays to confirm its novel mechanism of action in blocking NOTCH signaling. In addition, its anti-tumor activity was confirmed using xenograft models of NOTCH dependent human leukemias, breast cancer and PDX models of human T-ALL.

SUMMARY

CB-103 has shown the ability to block NOTCH signaling in human T cell acute lymphoblastic leukemia cancer cell lines and inhibits NOTCH dependent cellular processes in mice. Due to its novel mechanism of action, CB-103 exhibits anti-tumor efficacy in GSI resistant T-ALL cell lines. CB-103 also induces anti-tumor effect in a xenograft model of human triple negative breast cancer resistant to GSIs and Mabs against NOTCH ligands/receptors.

Furthermore, CB-103 has shown remarkable activity in *ex vivo* and *in vivo* patient derived models of human T-ALL harbouring activation of the NOTCH pathway.

CONCLUSIONS

Based on SAR and *in vivo* pharmacokinetic/ADME data, CB-103 and its analogs are undergoing additional preclinical studies to nominate a development candidate for clinical development.

Contact

Cellestia Biotech AG, Basel, Switzerland.

www. Cellestiabiotech.com

Email: raj.lehal@cellestiabiotech.com

REFERENCES

1. Andersson ER, Lendahl U. 2014. Therapeutic modulation of NOTCH signaling-are we there yet ? Nature reviews Drug discovery **13**: 357-378.
2. Andersson ER, Sandberg R, Lendahl U. 2011. NOTCH signalling: simplicity in design, versatility in function. Development **138**: 3593-3612.
3. Artavanis-Tsakonas S, Rand MD, Lake RJ.1999. NOTCH signaling: cell fate control and signal integration in development. Science **284**:770-776